Nilotinib Compassionate Use in Advanced GIST- A Retrospective Analysis

M. Montemurro,[1] P. Schöffski,[2] D. Pink,[3] H. Gelderblom,[4] H. Joensuu,[5] J. Schütte,[6] R. von Moos,[7] C. M. Wendtner,[8] B. Seddon,[9] S. Leyvraz[1]

University Hospital, Lausanne, Switzerland;[1] Leuven Cancer Institute, Leuven, Belgium;[2] Helios Hospital, Bad Saarow, Germany;[3] Leiden University Medical Center, Leiden, The Netherlands;[4] Helsinki University Hospital, Helsinki, Finland;[5] Marien Hospital, Düsseldorf, Germany;[6] Kantonsspital Graubünden, Chur, Switzerland;[7] University Hospital, Köln, Germany;[8] University College Hospital, London, United Kingdom[9]

INTRODUCTION

Tyrosine kinase inhibitors (TKI) have considerably improved outcome of patients with advanced gastrointestinal stromal tumors (GIST) and extended overall survival to more than five years. While median progression-free survival is approximately two years with first-line imatinib and 24 weeks with second-line sunitinib, treatment alternatives that may result in better outcomes are needed. Nilotinib is a second-generation TKI with comparable inhibitory activity as imatinib. A Phase I study has shown that nilotinib monotherapy has clinical activity in GIST.

BACKGROUND

GIST

- GIST are rare, mesenchymal neoplasms that arise from the interstitial cells of Cajal or their precursors
- GIST express the CD117 antigen – an epitope of KIT – a TK receptor
- Pathogenetic mutations code for a constitutively activated, mutant receptor (KIT, PDGRFα); oncogenic signaling occurs through the TK of the mutated receptor
- TKIs block the activity of KIT/PDGFRα and interrupt oncogenic signaling
- TKIs have considerably improved the outcome of advanced GIST
 - First-line imatinib—mOS 51–57 months, mPFS 18–20 months[1,2]
 - Second-line sunitinib—mOS 73 weeks, mPFS 24 weeks[3,4]

mOS=median overall survival; mPFS=median progression-free survival

Nilotinib (formerly known as AMN107)

- A second-generation, oral TKI engineered to have specific inhibitory activity against KIT, PDGFRα, and BCR-ABL
- Nilotinib achieves intracellular concentrations 7–10x greater than does imatinib in GIST cell lines[5]
- Maintains activity in imatinib-resistant GIST cell lines[6]
- Nilotinib monotherapy was found to have clinical activity in a recent Phase I study[7]
 - Study patients were imatinib-resistant/intolerant
 - 18 patients received nilotinib monotherapy (2 x 400 mg)
 - 14 patients (78%) obtained a clinical benefit (PR+SD)
 - mPFS was 168 days

PR=partial response; SD=stable disease

Figure 1. Nilotinib Chemical Structure

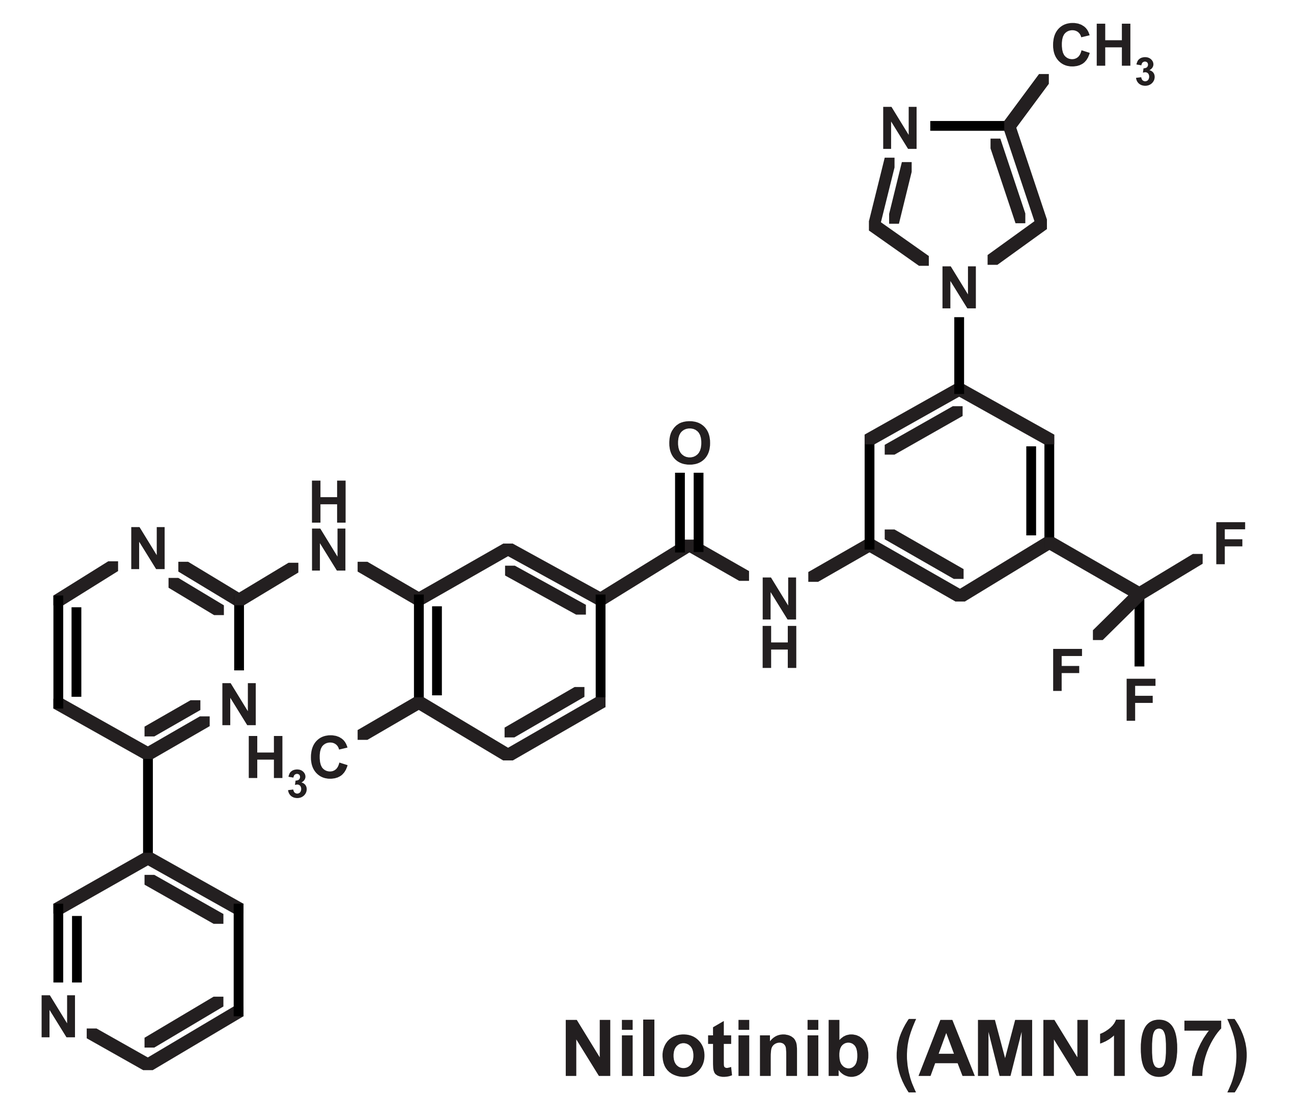

Nilotinib (AMN107)

OBJECTIVES OF STUDY

To assess the efficacy of the second-generation TKI, nilotinib, in a compassionate-use program for patients with no other available treatment options, including surgery and investigational agents, and who had failed prior TKI-therapy comprised of imatinib and sunitinib.

METHODS

Study Design

- Patients failing all available therapeutic options had access to nilotinib on a compassionate-use program. The starting dose of nilotinib was 400 mg twice daily (bid) with allowance for dose reduction to 400 mg once daily (qd) for toxicity
- Retrospective analysis on >1/3 of all patients treated with nilotinib compassionate use in Europe
- 12 centers in six European countries participating; all consecutive patients of each center were included

Program Statistics

Compassionate use was approved in 69 countries; 23 of these were in Europe
World: number of GIST approvals/requests: 352/384
Europe: number of GIST approvals/requests: 156/165

Inclusion Criteria

- Age ≥18 years
- WHO Performance: 0–2
- Histologically confirmed GIST, unresecteable and/or metastatic, not amenable to surgery or combined modality with curative intent
- Failure of all treatment options, including imatinib (up to 800 mg) and sunitinib
- Imatinib, sunitinib, investigational agents discontinued ≥5 days prior to start of nilotinib
- Normal organ function, electrolyte, and marrow function
- Written informed consent

Exclusion Criteria

- Any investigational drug ≤4 weeks prior to nilotinib
- Impaired cardiac function
- QT-interval prolonging medication and CYP3A4 inhibitors
- Severe and/or uncontrolled concurrent disease that, in the investigator's opinion, could cause unacceptable risks or compromise compliance
- Pregnancy; breast feeding; no contraception
- Major surgery ≤2 weeks prior to Visit 1 or not recovered from surgery
- Wide field radiotherapy ≤4 weeks or limited field radiation <2 weeks prior to Visit 1 or not recovered from such therapy
- Known ongoing alcohol or drug abuse
- Unwillingness or inability to comply with the treatment protocol

Baseline Patient Characteristics (Table 1)

- All patients had failed imatinib and sunitinib and all other available therapeutic options
- Mutational status was also determined for nearly half of study patients
- Study patients underwent a range of prior treatments including surgery and investigational agents, in addition to imatinib and sunitinib. Prior treatment details, including reason for treatment cessation with imatinib and sunitinib, are provided in Table 2.

Table 1. Patients' Baseline Characteristics

Characteristic	Patients (N=49)
Age	
Median age, years (range)	58 (24–80)
Sex	
Male:Female	34:15
Median follow-up, days (range)	176 (12–947)
At initial diagnosis, n	47
Localized	17
Advanced	7
Metastatic	23
Primary localization, n	42
Gastric	17
Small Bowel	17
Other	8
Mutational data, n	24
KIT exon 11	10
Wild-type	4
KIT exon 9	3
Other	7

Table 2. Prior-Treatment Baseline Characteristics

– Specifically with Imatinib and Sunitinib

Patients	Imatinib (n=49)	Sunitinib (n=49)
Treatment duration: median weeks (range)	139 (8–304)	24 (3–117)
Reason for cessation, n	49	47
Progressive disease	46	43
Toxicity	2	2
Progressive disease + toxicity	1	2

– With Other Treatments Prior to Nilotinib

Treatment		
Surgery, n (range)	1 (0–8)	
Investigational therapies	4	
AMG706*, later imatinib/everolimus, n	1	
AMG706, n	1	
PTK787†, n	2	

*AMG706, a multi-kinase inhibitor
†PTK787, an antiangiogenic, VEGFR inhibitor

RESULTS

Clinical benefit with nilotinib was observed in 45% of patients (8% PR + 37% SD) as shown in Figure 2.

Figure 2. Clinical Benefit in 45% of Patients Treated with Nilotinib

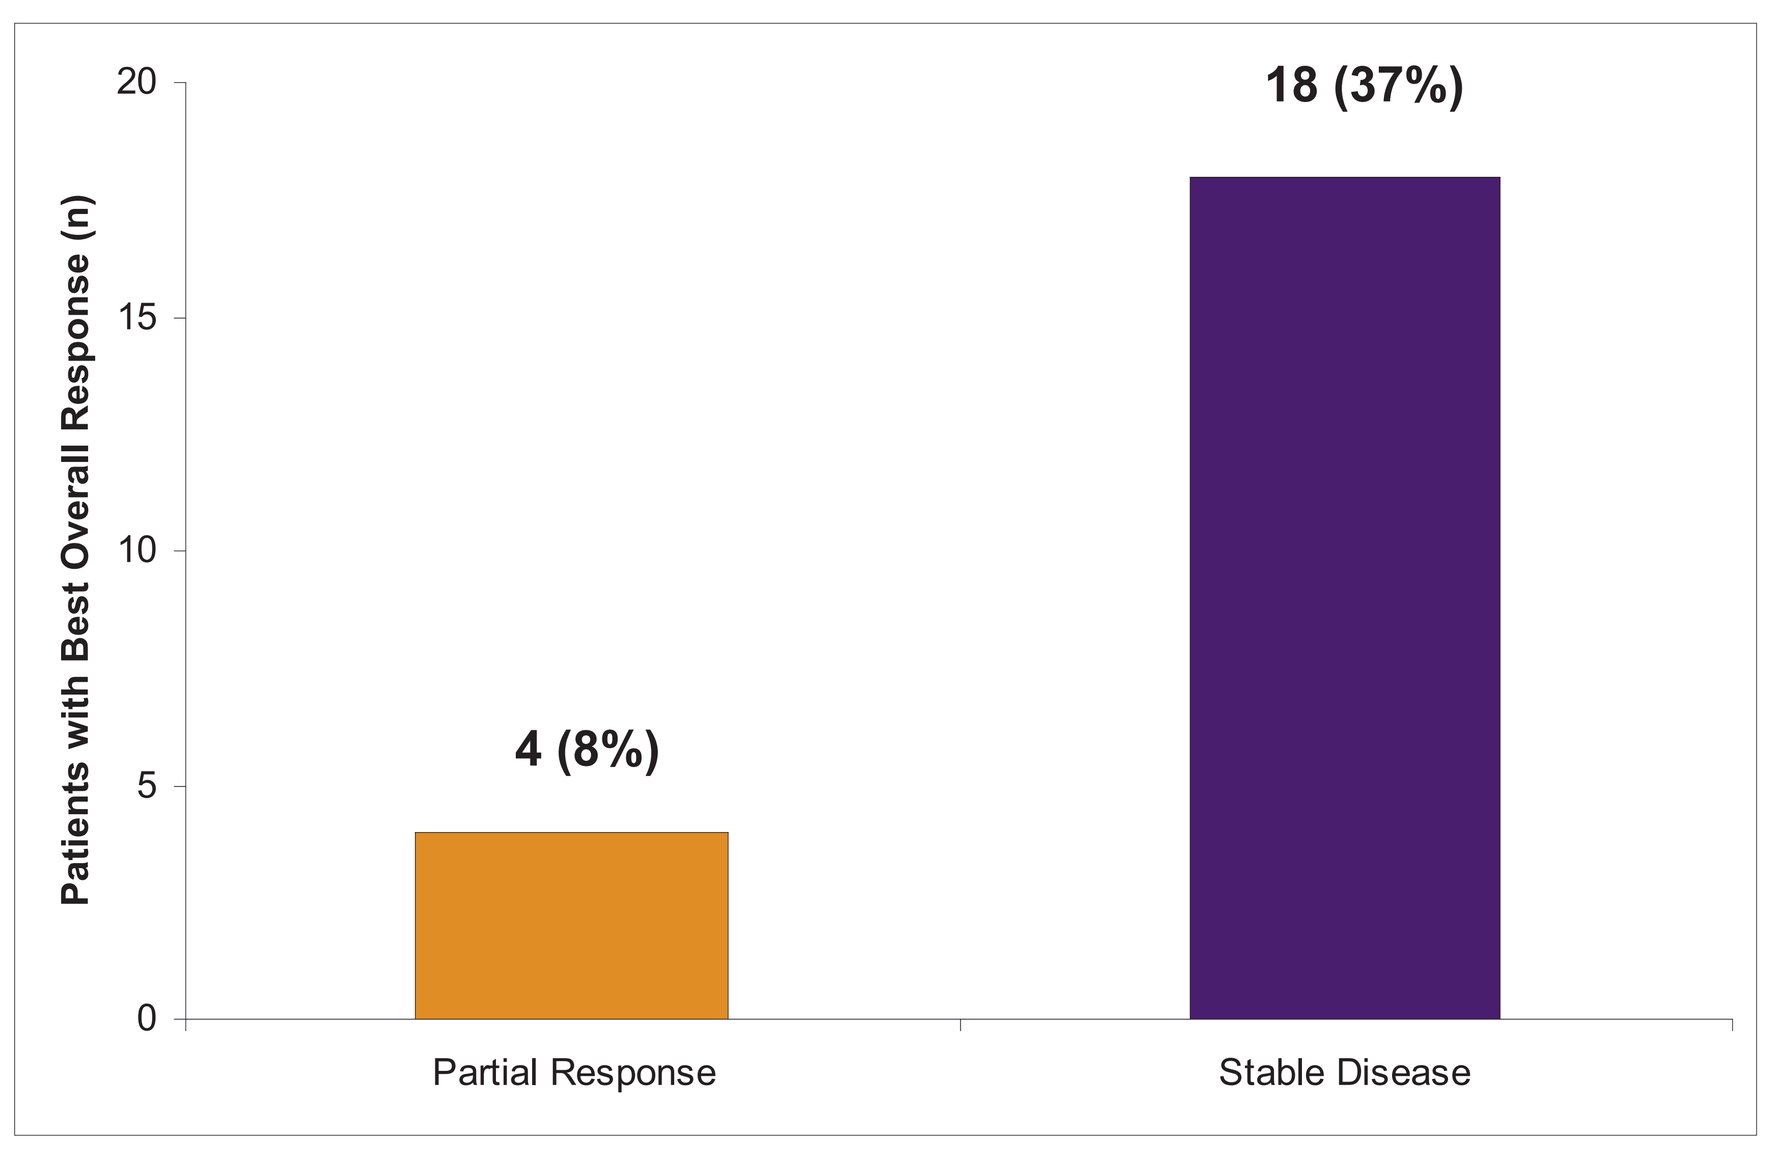

Patients with Best Overall Response (n)
0
5
10
15
20
4 (8%)
18 (37%)
Partial Response
Stable Disease

Treatment Duration

>2 months in 27 patients (55%)
>4 months in 16 patients (33%)
The median OS for patients on nilotinib is 263 days as shown in Figure 3

Figure 3. Overall Survival (OS) Since Nilotinib Start

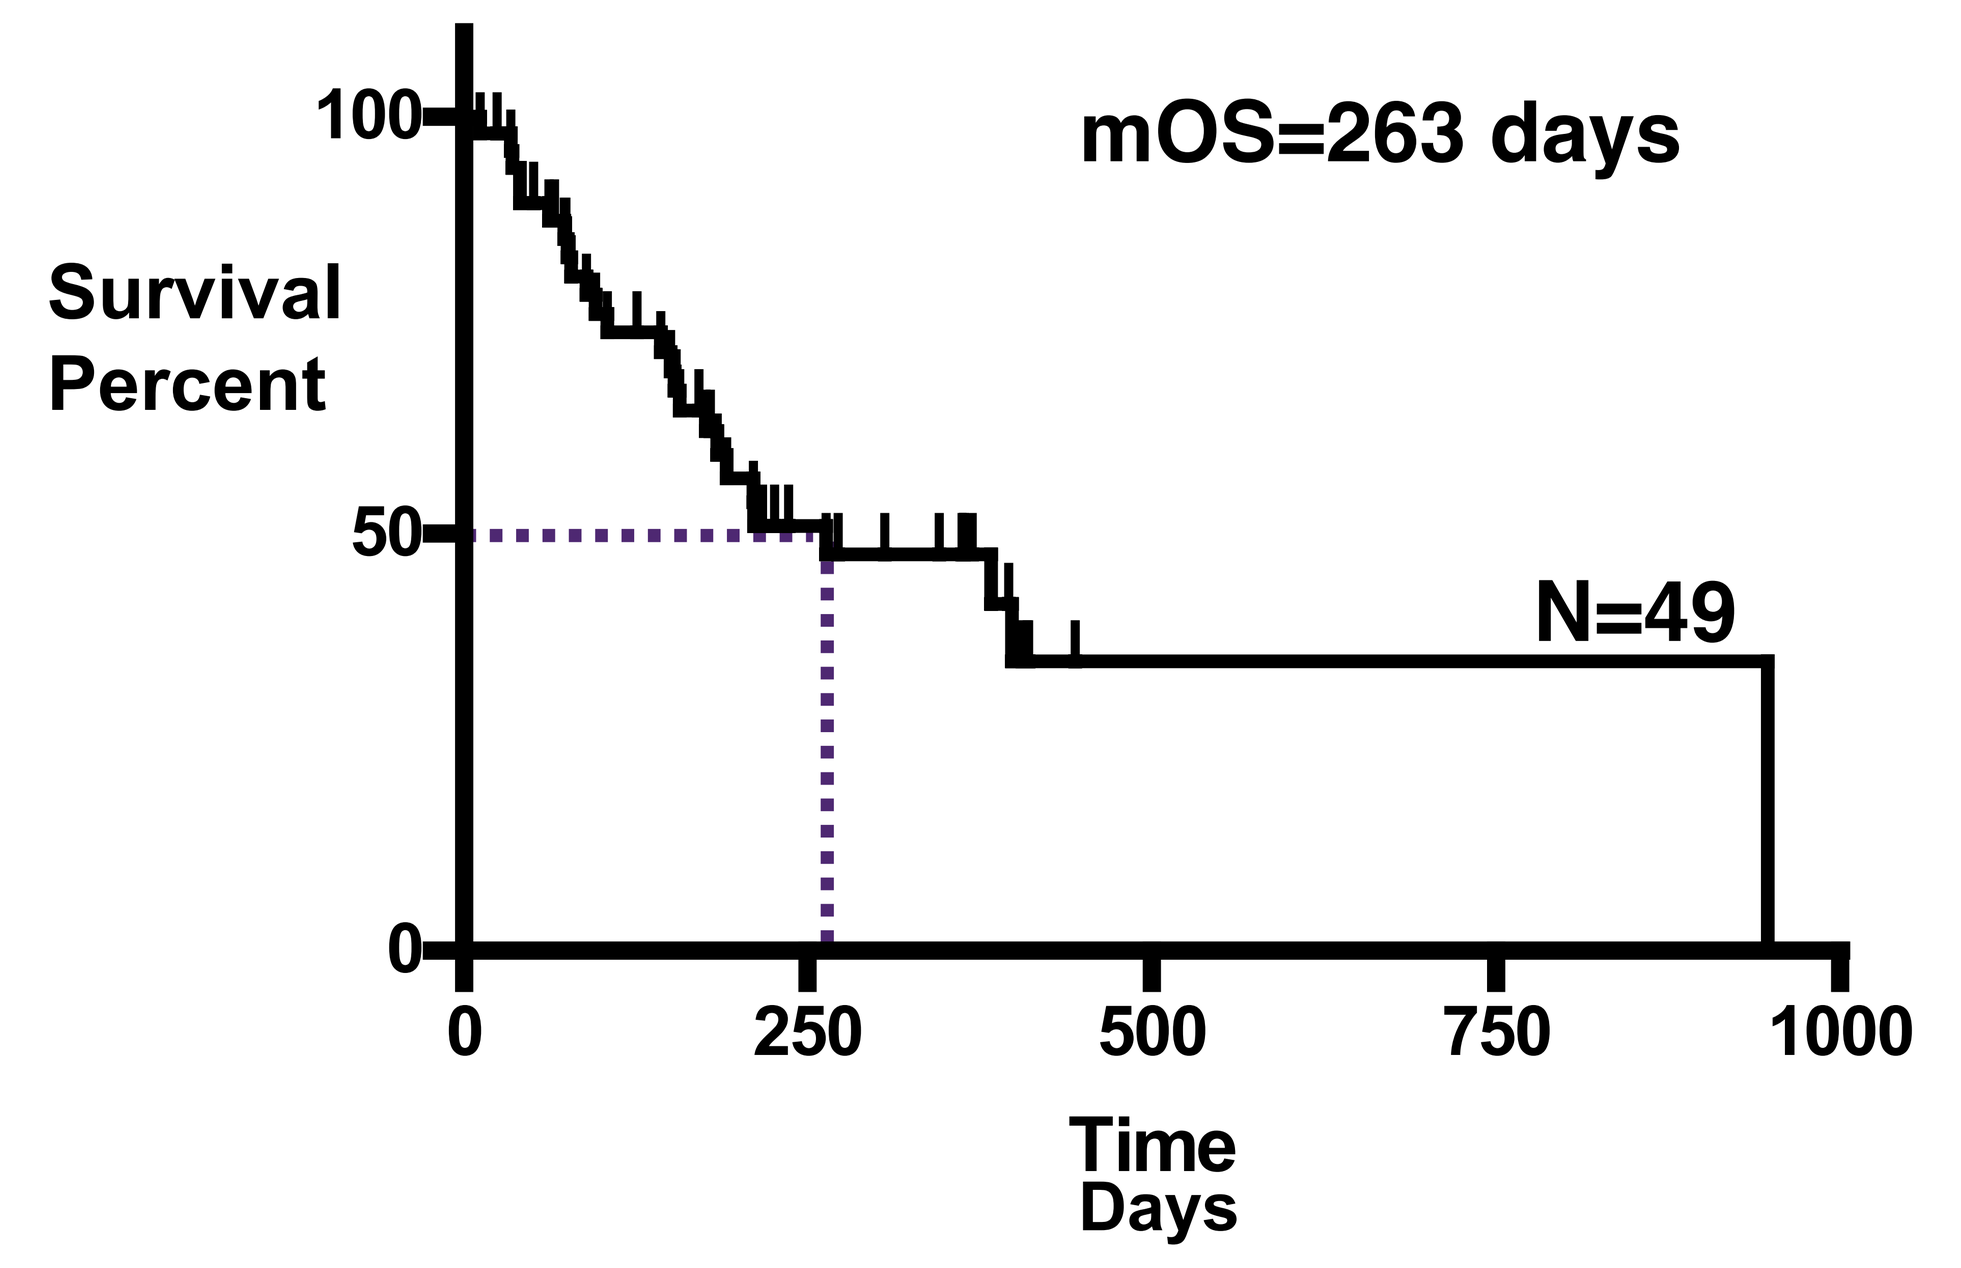

mOS=263 days
Survival Percent
100
50
0
N=49
0
250
500
750
1000
Time Days

Safety

Treatment was discontinued in six patients (12%) due to the following reasons:

- Nausea/anorexia
- Asthenia/anorexia/nausea/vomiting
- Muscle aches/fatigue/diarrhea
- Chest pain (cardiac)
- QT prolongation

CONCLUSIONS

This is the largest series reported to date that assessed the efficacy of nilotinib, a second-generation TKI, in patients with imatinib- and sunitinib-refractory GIST. Nilotinib was well tolerated and displayed clinical activity in this heavily pretreated group of GIST patients.

- Clinical benefit (PR+SD) in 45% of the patients
- Median overall survival (mOS) with nilotinib was 263 days

These results warrant further investigation of nilotinib in GIST, including its use in first- or second-line treatment.

REFERENCES

1. Blanke CD, Demetri GD, von Mehren M, et al. Long-term results from a randomized Phase II trial of standard- versus higher-dose imatinib mesylate for patients with unresectable or metastatic gastrointestinal stromal tumors expressing KIT. J Clin Oncol 2008;26:620-5.
2. Blanke CD, Rankin C, Demetri GD, et al. Phase III randomized, intergroup trial assessing imatinib mesylate at two dose levels in patients with unresectable or metastatic gastrointestinal stromal tumors expressing the kit receptor tyrosine kinase: S0033. J Clin Oncol 2008;26:626-32.
3. Morgan JA, Reichardt P, Kang YK, et al. Sunitinib (SU) in a worldwide treatment-use trial of patients with GIST: Safety and efficacy. ASCO Gastrointestinal Cancers Symposium 2008: Abstract No 31.
4. Demetri GD, van Oosterom AT, Garrett CR, et al. Efficacy and safety of sunitinib in patients with advanced gastrointestinal stromal tumour after failure of imatinib: a randomised controlled trial. Lancet 2006;368:1329-38.
5. Prenen H, Guetens G, de Boeck G, et al. Cellular uptake of the tyrosine kinase inhibitors imatinib and AMN107 in gastrointestinal stromal tumor cell lines. Pharmacology 2006;77:11-6.
6. Dileo P, Bauer S, Van Den Abbeele A, et al. Results with AMN107, a novel kinase inhibitor, in gastrointestinal stromal tumor (GIST): Preclinical rationale and early results in a patient (Pt) with imatinib (IM)-resistant GIST. ASCO Gastrointestinal Cancers Symposium 2006: Abstract No 53.
7. Blay JY, Casali PG, Reichardt P, et al. A phase I study of nilotinib alone and in combination with imatinib (IM) in patients with imatinib-resistant gastrointestinal stromal tumors (GIST) - Study update. ASCO Meeting Abstract 2008;26:10553.

ACKNOWLEDGEMENTS

- Switzerland — Arnaud Roth, Valerie Elsig, Sigrid Welte
- US — Mona Moonis, Jenna Fan, Mary-Anne Cox, Jim Wang, Richard Woodman
- Germany — Peter Reichardt, Simone Micheel, Lars Pester, Linda Heinze, Viktor Grünwald
- Belgium — Lieve Ons